INFLUENCE OF AN ACID ENVIRONMENT ON THE RELEASE BEHAVIOUR OF PELLETS CONTAINING PAPAVERINE

V. Daza[1], A. Aguilar de Leyva[2], V. Jiménez[2], L. M. Melgoza[3], I. Caraballo[2]

[1] Cronofarma SLL. General Armero, 52. 41420–Fuentes de Andalucía. Seville. Spain.
[2] Department of Pharmacy and Pharmaceutical Technology, University of Seville, C/Profesor García González, 2. 41012 - Seville, Spain.
[3] Departamento de Sistemas Biológicos, Universidad Autónoma Metropolitana-Xochimilco, Calzada del Hueso 1100, México D.F

Introduction

The solubility of a drug is a very important factor influencing its gastrointestinal absorption from oral dosage forms.
Papaverine hydrochloryde is a benzylisoquinoline alkaloid which has a direct and non-specific relaxant effect on vascular, cardiac, and other smooth muscle. Its potent vasodilator effect has led to its widespread clinical use in the treatment of vasospasmic diseases such as cerebral spasm associated with subarachnoid hemorrhage (Fandino et al., 1998). Papaverine hydrochloryde is a salt of a basic drug which shows a low and pH dependent solubility in water. For this reason, this drug shows important bioavailability problems (Kraus et al., 1991), especially when it is included in sustained release formulations. Organic acids can be added to the formulations of papaverine hydrochloryde. These acids act as pH modifiers, contributing to the ionization of basic drugs and thus enhancing its solubility.
Pellets are multiparticulate systems which offer many therapeutic and technological advantages compared with monolithic systems: they disperse as individualized units in the gastrointestinal tract reducing high local drug concentration, maximizing drug absorption and reducing peak plasma fluctuation. Another additional therapeutic advantage is that these dosage forms eliminate the dependence of the drug effect on gastric emptying, reducing intra and interindividual variability of the drug plasma concentrations (Bodmeier, 1997). In order to overcome bioavailability problems of pellets containing low soluble drugs, coated pellets using a functional core instead of an inert core can be manufactured (Xu et al., 2011).

Objectives

❑ To investigate the influence of an acid microenvironment, obtained with tartaric acid core pellets (TAP™) on the release profiles of papaverine hydrochloryde from controlled release coated pellets prepared with a Surelease™ layer of varying thicknesses.
❑ To compare their release profiles with pellets prepared using standard sugar starter cores (Suglets™).

Experimental methods

Materials:
✓Standard sugar cores (Suglets™) (Colorcon, Spain),
✓ Tartaric acid cores (TAP™) (Pharmatrans Sanaq, Switzerland)
✓ Papaverine·HCl (Acofarma, Spain)
✓HPMC (Colorcon, UK)
✓Opadry™ (Colorcon, UK)
✓Ethylcellulose based suspension (Surelease™) (Colorcon, UK).

Table 1: structure of the pellet formulations prepared.

	Components
Core	TAP™
Layer 1	HPMC (protective shell)
Layer 2	Papaverine·HCl + Binder
Layer 3	Surelease™ (controlled release shell)

Table 1: Multilayer structure of the coated pellets prepared.

Preparation of the coated pellets:
❑ Fluidised bed system (Innojet, Ventilus-1) employing different percentages of surelease™ (10%, 15%, 16.6%, 20% and 25% w/w). The following conditions have been employed for the coating process:

Layer	Air flow (m^3/h)	Temperature (ºC)	Coating Speed (g/min.)
1	55	45	0.68
2	55	44	0.68
3	60	40	0.60

Dissolution studies:
❑ USP Dissolution apparatus Sotax AT7 smart (Allschwil, Switzerland):
- basket method
- three replicates
- 900 ml of pH 6.8 buffer at 37±0.5ºC
- 50 rpm.

❑ UV/VIS Spectrophotometer Agilent 8453 (California, USA). The percentage of drug released was measured in a at a wavelength of 250 nm.

Results and discussion

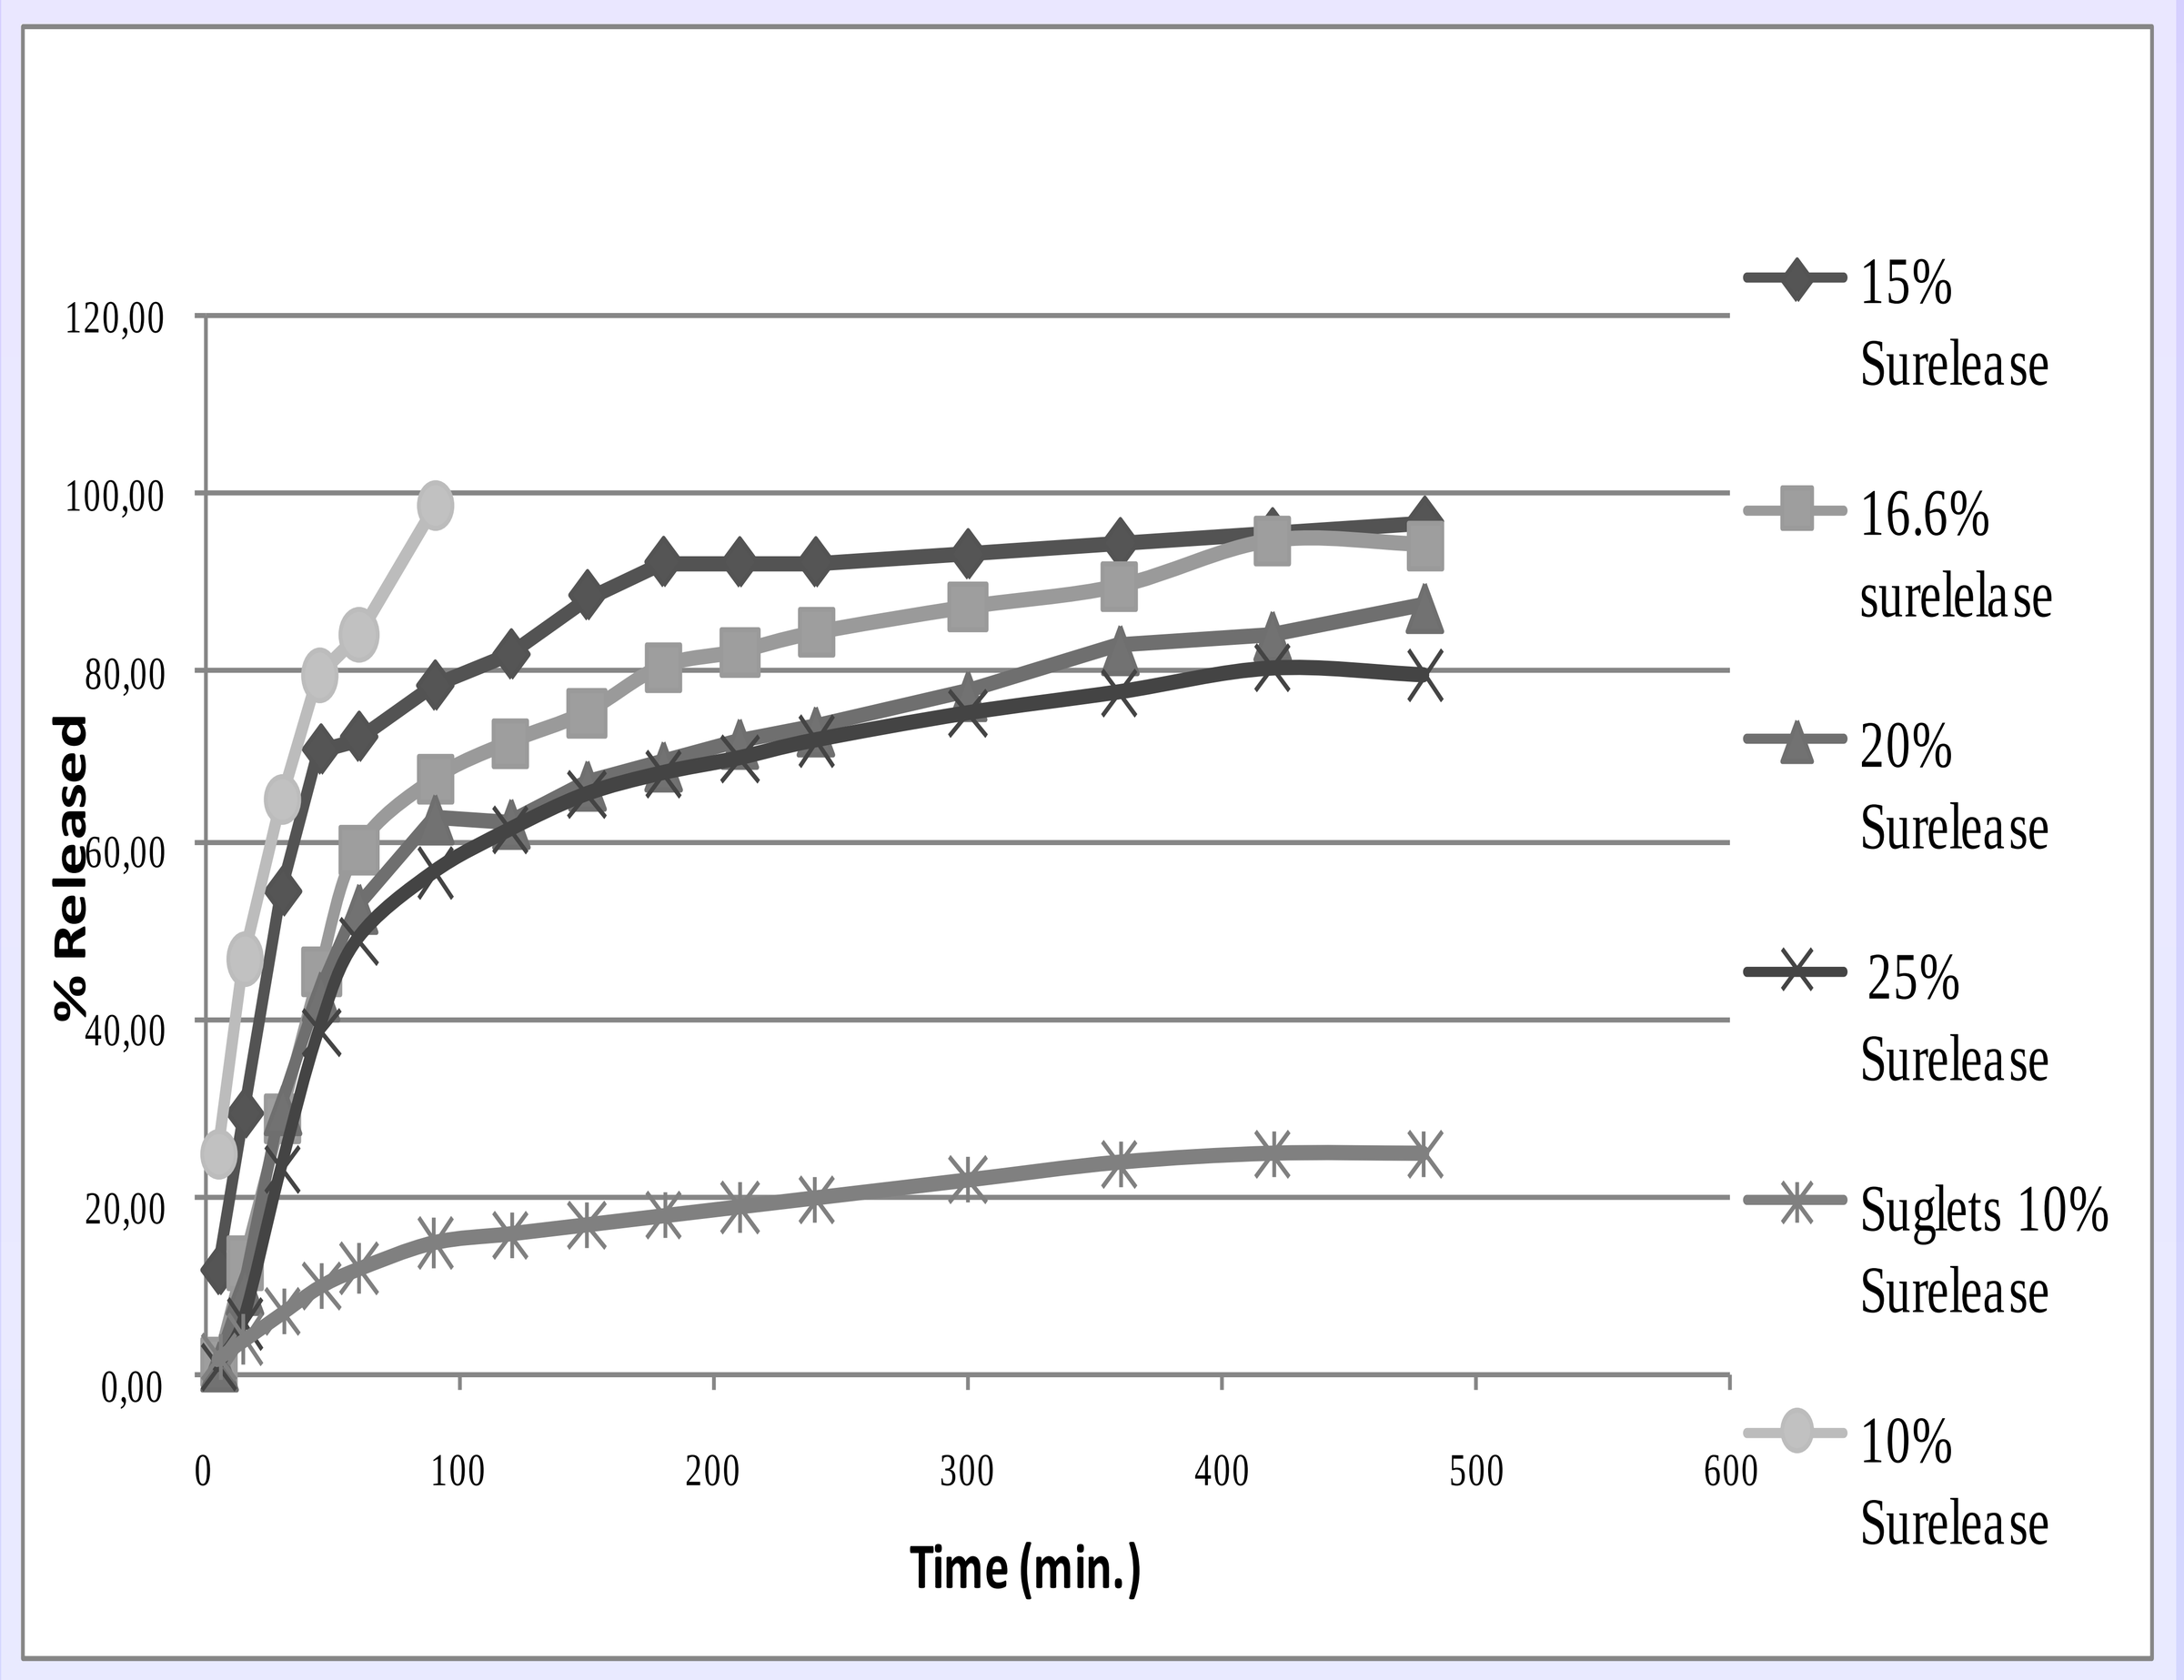
120,00
100,00
80,00
60,00
40,00
20,00
0,00
% Released
0
100
200
300
400
500
600
Time (min.)
15% Surelease
16.6% surelelase
20% Surelease
25% Surelease
Suglets 10% Surelease
10% Surelease

Figure 1: Release profiles of the coated pellets prepared

Figure 1 shows the release profiles of the coated pellets prepared using TAP™ or Sugar pellets as initial cores.
It can be observed that while the percentage of papaverine hydrochloryde released from pellets with sugar cores and 10% of Surelease™ is lower than 30% after 8 hours, pellets containing tartaric acid cores and 10% of Surelease™ released 100% of papaverine hydrochloryde in approximately 90 minutes.
It is also clear from this figure that an increase in the percentage of Surelease™ (which supposes an increase in the thickness of layer 3) results in a stronger control of the release of Papaverine hydrochloride. This allows modulating the release rate from the obtained pellets.

Conclusions

Using tartaric acid pellets (TAP™) as starting cores, the acidic pH inside the pellets makes possible to have a complete release of Papaverine·HCl. This is expected to **improve markedly the bioavailability of Papaverine hydrochloryde as well as of basic drugs with low solubility in neutral pH**.
This strategy allows **overcoming the difficulty** of preparing a **controlled release** dosage form containing papaverine·HCl.

Aknowledgements

The authors wish to acknowledge Pharmatrans Sanaq for supplying TAP™ and Colorcon for supplying Suglets™.

References

1. Fandino, J., et al., Improvement of cerebral oxygenation patterns and metabolic validation of superselective intraarterial infusion of papaverine for the treatment of cerebral vasospasm. J. Neurosurg., 89, 93–100. (1998).
2. Kraus, C., et al., Pharmacokinetics and bioavailability of papaverine HCl following intravenous, peroral, rectal, vaginal, topical and bucal administration in beagle dogs. Biopharm. Drug. Dispos., 12, 537-546. (1991).
3. Bodmeier, R., Tableting of coated pellets. Eur. J. Pharm. Biopharm., 43, 1–8. (1997).
4. Xu, L., et al., Preparation and in vitro–in vivo evaluation of none gastric resident dipyridamole (DIP) sustained-release pellets with enhanced bioavailability.In.J.Pharm. (2011),doi:10.1016/j.ijpharm.2011.10.014.